Peripheral Edema and Weight Gain in Adult Patients With Painful Diabetic Peripheral Neuropathy Receiving Gabapentin Enacarbil or Pregabalin Enrolled in a Randomized Phase 2 Trial

I6-1.004

Anne M. Calkins,[1] Joseph Shurman,[2] Mark Jaros,[3] Richard Kim,[4] Gwendoline Shang[4]

[1]New York Spine and Wellness Center, North Syracuse, NY; [2]Scripps Memorial Hospital La Jolla, CA; [3]Summit Analytical, LLC, Denver, CO; [4]XenoPort, Inc., Santa Clara, CA

Background

- Gabapentin enacarbil (GEn) is an actively transported prodrug of gabapentin that has been approved for the management of postherpetic neuralgia (1200 mg daily dose administered as 600 mg twice daily) and for the treatment of moderate-to-severe primary RLS in adults (600 mg once daily) by the United States FDA.[1] GEn is not approved for use in the treatment of diabetic peripheral neuropathy (DPN).
- In this phase 2 study of patients with neuropathic pain attributed to DPN, neither the GEn (1200 mg/day, 2400 mg/day, or 3600 mg/day) nor the control (pregabalin 300 mg/day, which is the FDA-approved dose for the treatment of DPN) arms differed from placebo with regard to the primary efficacy endpoint (change from baseline in the mean 24-hour average pain intensity score).
 - Differences in mean 24-hour average pain intensity score between GEn and placebo were: GEn 1200 mg/day, −0.35 (P=0.295); GEn 2400 mg/day, −0.02 (P=0.946); and GEn 3600 mg/day, −0.55 (P=0.105).
- In addition, GEn was well tolerated across all studied doses, including at the highest dose of 3600 mg/day.[2]
- As this was the first study to assess treatment with GEn compared with the approved dose of pregabalin, two treatment-related adverse events of clinical significance commonly reported with the alpha-2-delta class of drugs, peripheral edema and weight gain, were further examined.

Objective

- To examine the incidences of peripheral edema and weight gain in adult patients with painful DPN receiving GEn, pregabalin, or placebo in a phase 2 trial.

Methods

Study Design

- The design of this 20-week, phase 2, multicenter, randomized, double-blind, double-dummy, parallel-group, placebo-controlled, and active-controlled study has been previously published and is shown in Figure 1 (Clinicaltrials.gov NCT00643760).[2]
- Patients were randomized (2:1:1:1:2) to receive GEn 3600 mg/day, GEn 2400 mg/day, GEn 1200 mg/day, pregabalin 300 mg/day, or matching placebo.[2]

Patients

- Eligible patients were men and women ≥18 years of age, diagnosed with diabetes mellitus (type 1 or 2) who had pain attributed to DPN, defined as painful distal symmetric sensorimotor polyneuropathy.[2]
- Patients had DPN for at least 6 months and no greater than 5 years; pain attributed to their DPN of at least moderate intensity (average pain intensity score of ≥4 on an 11-point pain intensity numerical rating scale where 0=no pain and 10=worst possible pain); stable glycemic control 3 months prior to randomization; and glycosylated hemoglobin (HbA1c) levels <8% at the time of randomization.[2]

Analyses

- The safety population included all randomized patients who received at least one dose of study medication. Overall safety and tolerability have been reported previously.[2]
- Safety assessments included treatment-emergent adverse events (TEAEs), body weight, and pedal edema. The incidences of each outcome were summarized by treatment group.
- The TEAEs of peripheral edema and weight gain were further explored in this analysis. In addition to the incidences of peripheral edema and weight gain by treatment groups, the proportions of patients with worsening pedal edema (worsening=positive change in grade) and weight gain ≥7% at any post-randomization visit were summarized, as well as mean changes in body weight from baseline.

Figure 1. Study Design

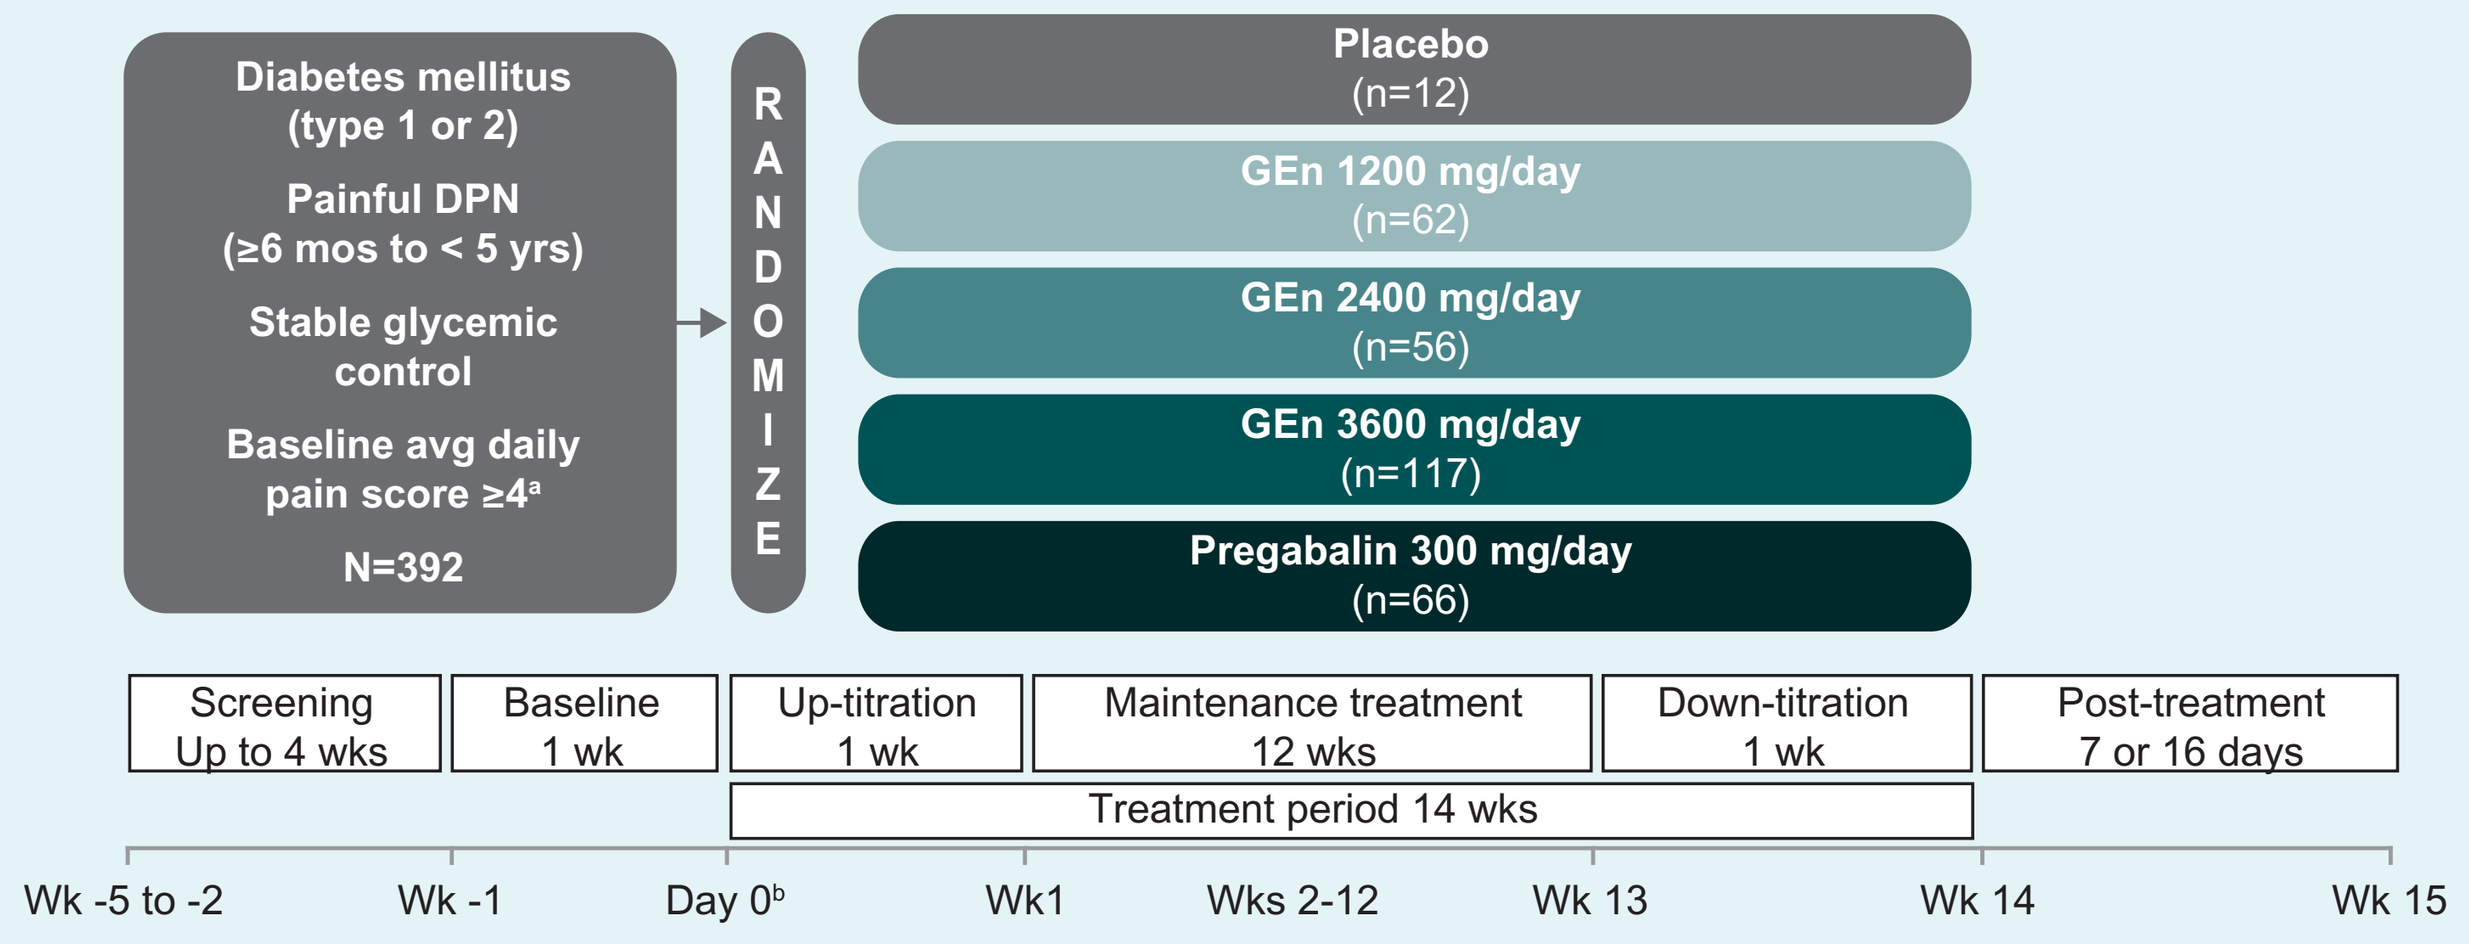
Diabetes mellitus (type 1 or 2)
Painful DPN (≥6 mos to < 5 yrs)
Stable glycemic control
Baseline avg daily pain score ≥4[a]
N=392
RANDOMIZE
Placebo (n=12)
GEn 1200 mg/day (n=62)
GEn 2400 mg/day (n=56)
GEn 3600 mg/day (n=117)
Pregabalin 300 mg/day (n=66)
Screening Up to 4 wks
Baseline 1 wk
Up-titration 1 wk
Maintenance treatment 12 wks
Down-titration 1 wk
Post-treatment 7 or 16 days
Treatment period 14 wks
Wk -5 to -2
Wk -1
Day 0[b]
Wk1
Wks 2-12
Wk 13
Wk 14
Wk 15

[a]On an 11-point pain intensity numerical rating scale (0=no pain; 10=worst possible pain).
[b]Randomization occurred at Day 0.

DPN = diabetic peripheral neuropathy; GEn = gabapentin enacarbil; Mos = months; Wk = week; Yrs = years.

Results

- A total of 421 patients were randomized to the treatment arms. One patient in the GEn 3600-mg group was randomized in error, did not take study medication, and therefore was excluded from the safety population.[2]
- Demographic and baseline characteristics are shown in Table 1.

Table 1. Baseline Characteristics of the Safety Population[a]

Characteristic	Placebo (n=120)	GEn 1200 mg (n=62)	GEn 2400 mg (n=56)	GEn 3600 mg (n=116)	Pregabalin 300 mg (n=66)
Age, years					
Mean (SD)	60.1 (10.63)	57.5 (10.32)	60.8 (8.97)	57.5 (9.87)	57.7 (10.59)
Min-max	29–85	32–77	33–78	35–79	35–79
Age group, n (%)					
≤65 years	82 (68)	44 (71)	39 (70)	90 (78)	50 (76)
>65 years	38 (32)	18 (29)	17 (30)	26 (22)	16 (24)
Gender, n (%)					
Female	47 (39)	28 (45)	19 (34)	45 (39)	32 (48)
Male	73 (61)	34 (55)	37 (66)	71 (61)	34 (52)
Race, n (%)					
White[b]	98 (82)	46 (74)	47 (85)	90 (79)	52 (81)
BMI, n (%)					
≤30 kg/m²	27 (23)	16 (26)	22 (39)	39 (34)	17 (26)
>30 kg/m²	93 (78)	46 (74)	34 (61)	77 (66)	49 (74)
Baseline 24-hour pain score					
Mean (SD)	6.49 (1.26)	6.64 (1.47)	6.26 (1.22)	6.48 (1.43)	6.51 (1.27)
Min-max	4.0–10.0	4.0–9.4	4.0–9.6	4.0–10.0	4.0–9.3
Baseline 24-hour pain score, n (%)					
4 to <6.5	61 (51)	25 (40)	32 (57)	63 (54)	33 (50)
6.5 to 10	59 (49)	37 (60)	24 (43)	53 (46)	33 (50)
HbA1c					
Mean % (SD)	7.5 (1.3)	7.4 (1.3)	7.6 (1.4)	7.6 (1.4)	7.5 (1.3)

[a]421 patients were randomized to treatment; one patient in the GEn 3600 mg group did not take study medication and was excluded from the safety population; [b]Other races were primarily of African-American/African heritage.

BMI = body mass index; GEn = gabapentin enacarbil; HbA1c = hemoglobin A1c (glycated hemoglobin); SD = standard deviation.

Adverse Events

- Across treatment groups, the majority of patients reported at least one TEAE. The most common TEAEs that occurred in ≥5% of patients in either the GEn or pregabalin treatment groups included dizziness, somnolence, nausea, and peripheral edema (Table 2). All of the most common TEAEs occurred more frequently in at least one active treatment arm than in the placebo arm.[2]

Table 2. Most Common Treatment-Emergent Adverse Events Reported in >5% of the Safety Population[a]

Adverse event, n (%)	Placebo (n=120)	GEn 1200 mg (n=62)	GEn 2400 mg (n=56)	GEn 3600 mg (n=116)	Pregabalin (n=66)
Any event	79 (66)	45 (73)	38 (68)	86 (74)	47 (71)
Dizziness	7 (6)	9 (15)	8 (14)	16 (14)	9 (14)
Somnolence	5 (4)	2 (3)	7 (13)	14 (12)	9 (14)
Nausea	9 (8)	7 (11)	4 (7)	7 (6)	3 (5)
Peripheral edema	5 (4)	2 (3)	0 (0)	11 (9)	11 (17)
Headache	9 (8)	3 (5)	4 (7)	4 (3)	6 (9)
Muscle spasms	4 (3)	6 (10)	0 (0)	11 (9)	3 (5)
Diarrhea	6 (5)	3 (5)	2 (4)	6 (5)	5 (8)
Urinary tract infection	5 (4)	3 (5)	4 (7)	6 (5)	4 (6)
Constipation	4 (3)	3 (5)	4 (7)	4 (3)	6 (9)
Fatigue	3 (3)	3 (5)	3 (5)	5 (4)	4 (6)
Pain in extremity	2 (2)	1 (2)	4 (7)	6 (5)	2 (3)
Increased weight	1 (<1)	0 (0)	2 (4)	5 (4)	5 (8)
Dry mouth	4 (3)	0 (0)	4 (7)	1 (<1)	1 (2)

[a]421 patients were randomized to treatment; one patient in the GEn 3600 mg group did not take study medication and was excluded from the safety population.

GEn = gabapentin enacarbil.

- Compared with pregabalin, patients randomized to GEn experienced less peripheral edema and weight gain (Figures 2 and 3).

Figure 2. Incidence of Peripheral Edema in the Safety Population

A) Percentage of patients with peripheral edema as an adverse event

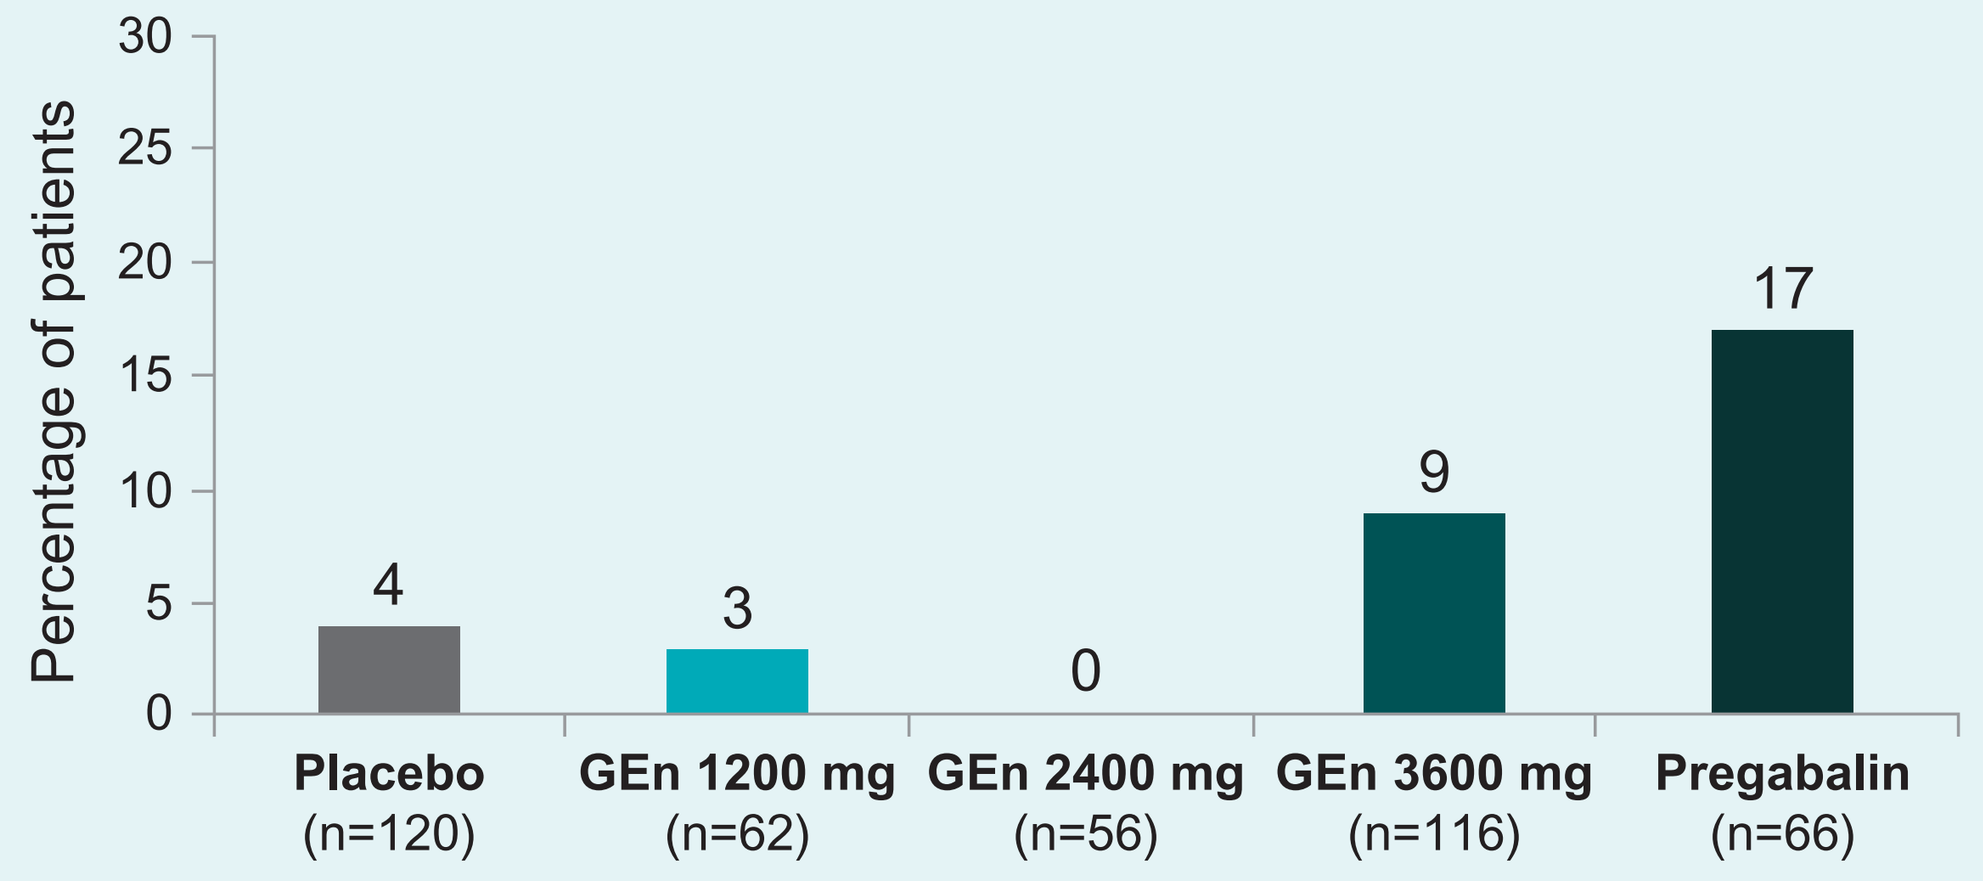
Percentage of patients
4
3
0
9
17
Placebo (n=120)
GEn 1200 mg (n=62)
GEn 2400 mg (n=56)
GEn 3600 mg (n=116)
Pregabalin (n=66)

B) Percentage of patients with worsening of pedal edema[a] relative to baseline

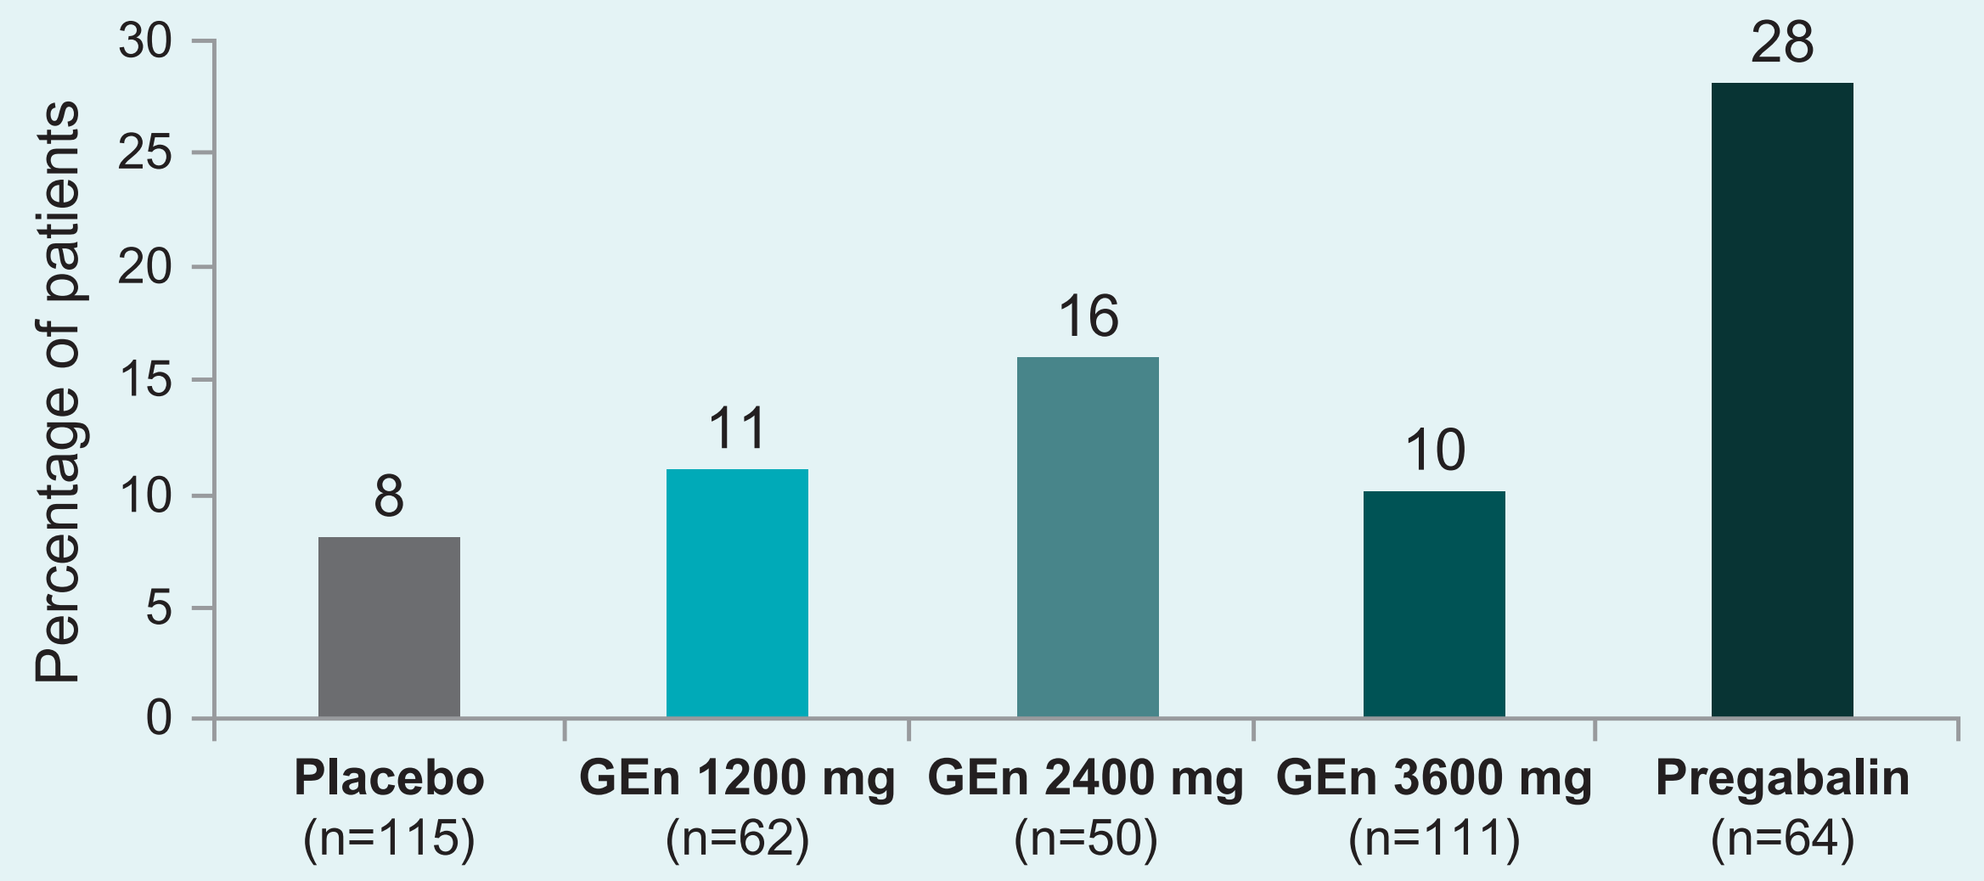
Percentage of patients
8
11
16
10
28
Placebo (n=115)
GEn 1200 mg (n=62)
GEn 2400 mg (n=50)
GEn 3600 mg (n=111)
Pregabalin (n=64)

[a]Worsening = positive change in grade; GEn = gabapentin enacarbil.

Figure 3. Incidence of Weight Gain in the Safety Population

A) Percentage of patients with weight gain as an adverse event

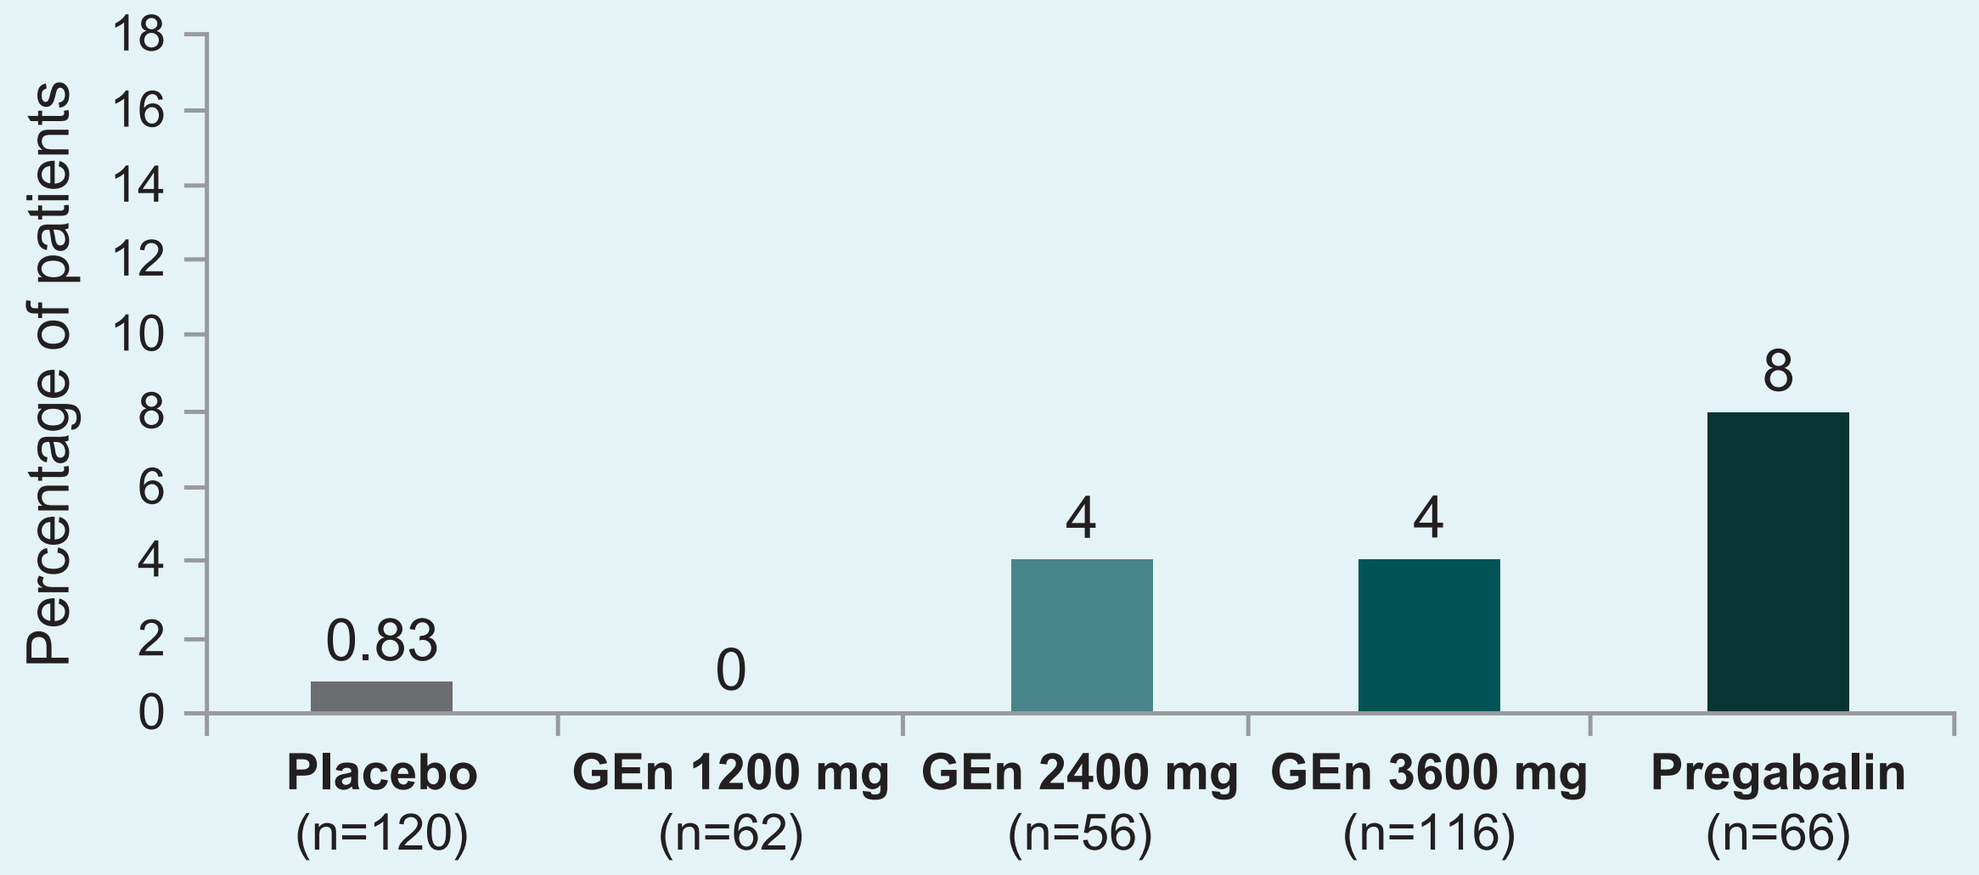
Percentage of patients
0.83
0
4
4
8
Placebo (n=120)
GEn 1200 mg (n=62)
GEn 2400 mg (n=56)
GEn 3600 mg (n=116)
Pregabalin (n=66)

B) Percentage of patients with ≥7% weight gain at any post-randomization visit

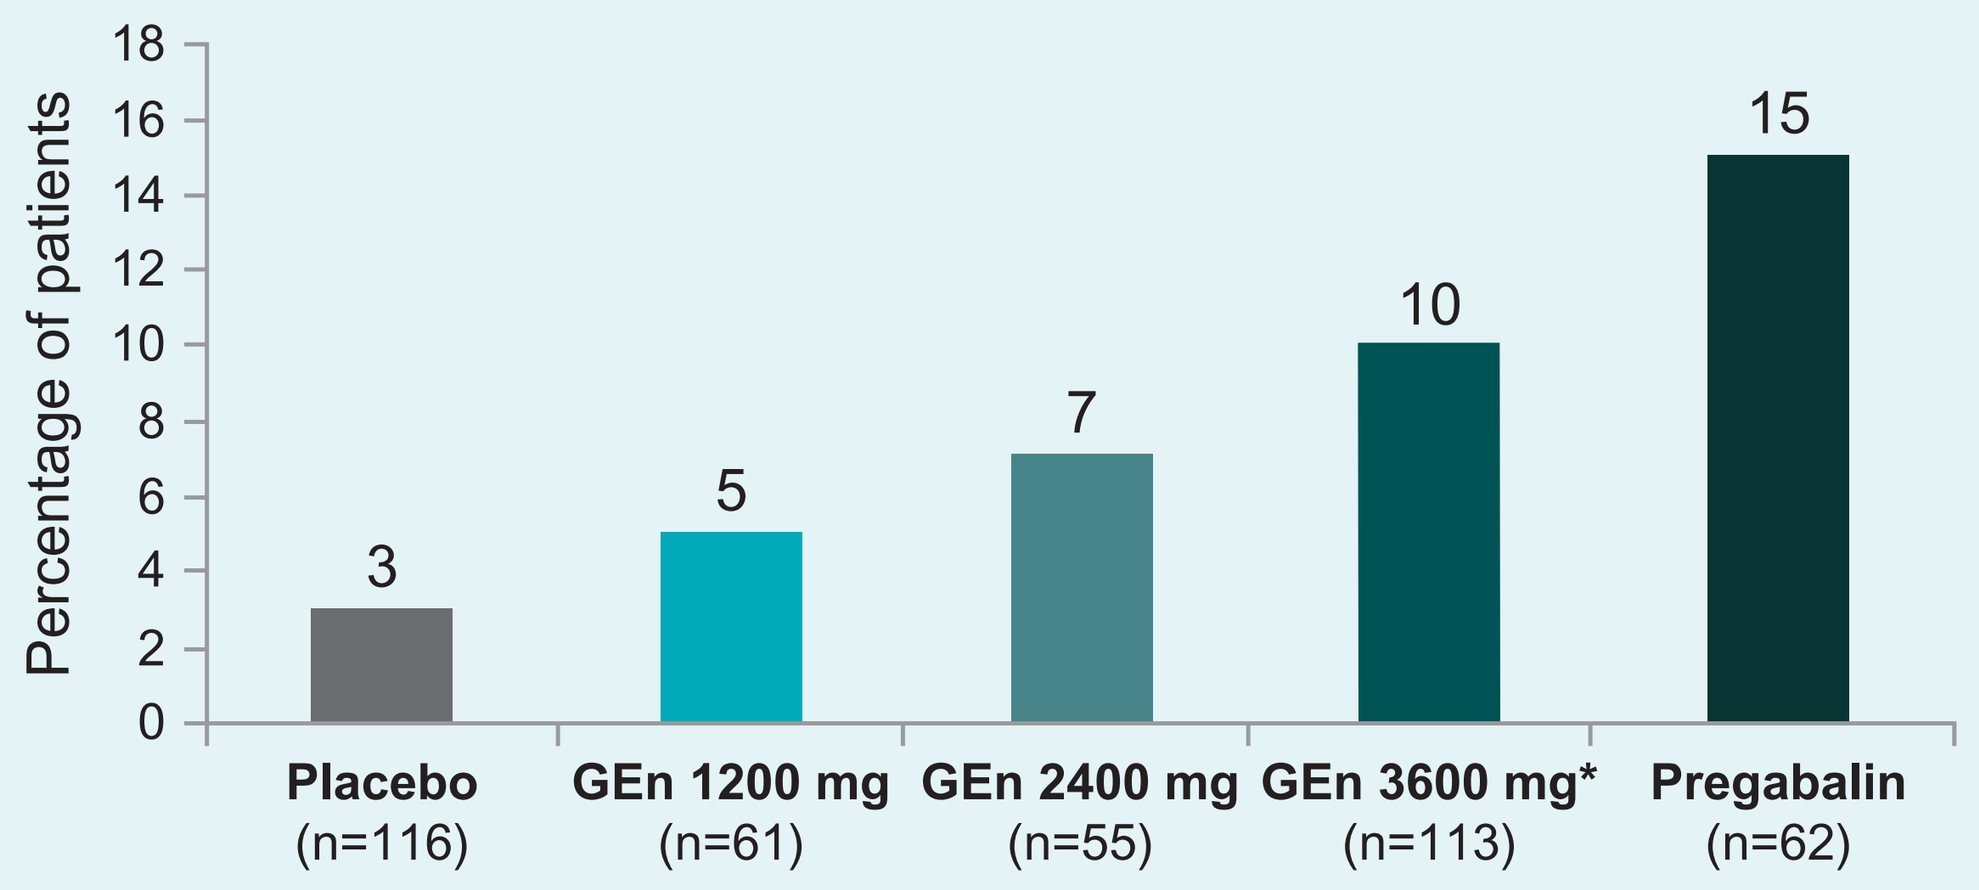
Percentage of patients
3
5
7
10
15
Placebo (n=116)
GEn 1200 mg (n=61)
GEn 2400 mg (n=55)
GEn 3600 mg* (n=113)
Pregabalin (n=62)

GEn = gabapentin enacarbil.
*One patient excluded due to implausible weight change data

- Figure 4 shows the mean weight changes from baseline at the end of week 13 on drug treatment phase or at time of early termination (LOCF). In the GEn treatment groups, weight gain appeared to increase with dose; patients in the 3600-mg group experienced the most weight gain and those in the 1200-mg group experienced the least weight gain.

Figure 4. Mean Change From Baseline in Weight Gain at week 13/ET (LOCF)*

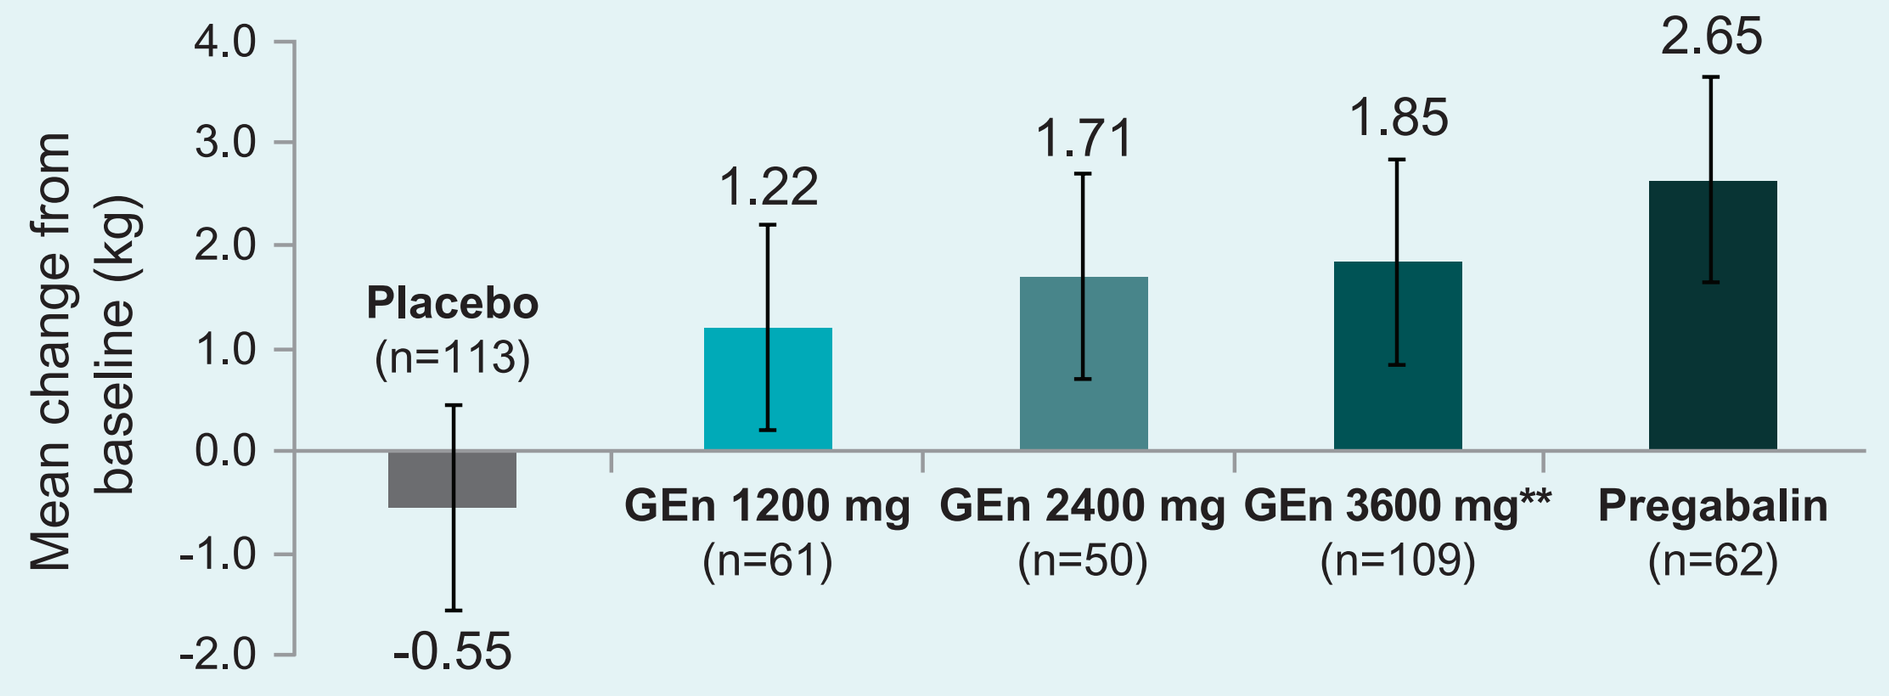
Mean change from baseline (kg)
Placebo (n=113)
-0.55
1.22
1.71
1.85
2.65
GEn 1200 mg (n=61)
GEn 2400 mg (n=50)
GEn 3600 mg** (n=109)
Pregabalin (n=62)

ET = early termination; LOCF = last observation carried forward; GEn = gabapentin enacarbil.
*Positive values correspond to weight gain; negative values correspond to weight loss.
**One patient excluded due to implausible weight change data.

- By week 5 (after 4 weeks of the maintenance treatment phase), patients in the pregabalin group had experienced a greater increase in weight from baseline (Figure 5). Mean body weight increased over time in all active treatment groups, although changes were smaller in the lower-dose GEn groups.

Figure 5. Mean Change From Baseline in Weight Gain[a]

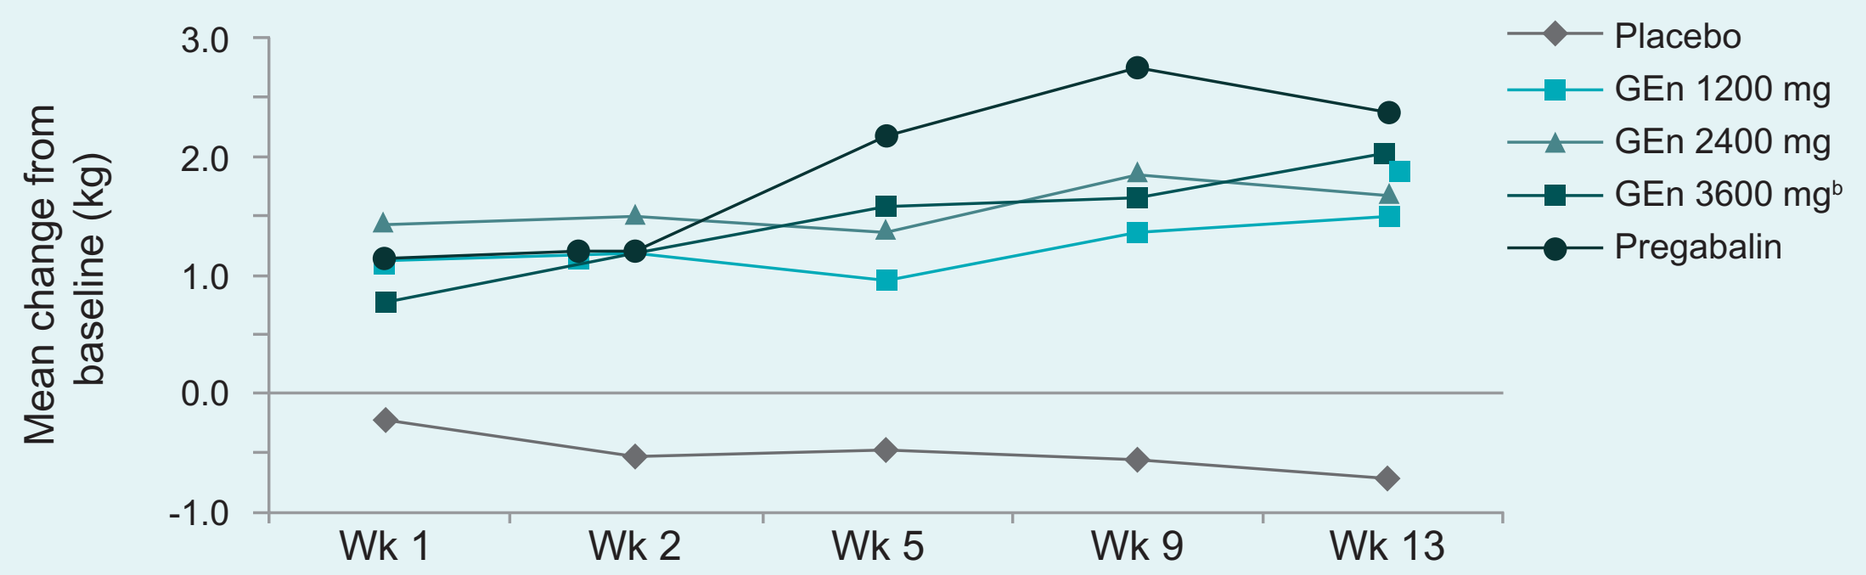
Mean change from baseline (kg)
Placebo
GEn 1200 mg
GEn 2400 mg
GEn 3600 mg[b]
Pregabalin
Wk 1
Wk 2
Wk 5
Wk 9
Wk 13

Patients, N	Wk 1	Wk 2	Wk 5	Wk 9	Wk 13
Placebo	112	108	103	97	90
GEn 1200 mg	61	58	57	51	48
GEn 2400 mg	49	48	43	40	39
GEn 3600 mg	109	106	96	84	81
Pregabalin	62	60	55	53	46

[a]Observed data. [b]One subject excluded due to implausible weight change data.
GEn = gabapentin enacarbil; W = week.

Conclusions

- In this phase 2 study of patients with painful DPN, treatment with GEn was associated with overall lower incidences of peripheral edema and weight gain compared with pregabalin.
 - This was most evident with GEn 1200 mg, but even the higher doses of GEn were associated with less weight gain and peripheral edema than pregabalin.
 - The weight gain in GEn-treated patients appeared to be dose-dependent.
- The observed differences in peripheral edema and weight gain between all GEn doses examined and pregabalin are of clinical significance, given that these TEAEs may lead to limitations in patients' willingness to tolerate and adhere to prescribed regimens.
- GEn is approved for the management of PHN in adults at a daily dose of 1200 mg. Additional benefit of using doses greater than 1200 mg/day was not demonstrated, and these higher doses resulted in an increase in adverse reactions.

References
1. Horizant [Prescribing Information]. Santa Clara, CA: XenoPort, Inc; 2013.
2. Rauck R, et al. *Pain Pract*. 2013;13:485–96.

Acknowldgements
The study was conducted by XenoPort, Inc., Santa Clara, CA. Financial support for medical writing was provided by XenoPort, Inc.

Disclosures
AC is a speaker for Depomed, Pfizer, Purdue, Salix, and Teva. JS has served as a consultant for Millennium, XenoPort, Insys Therapeutics, Purdue Pharma, Frederick, Teva Pharmaceutical Industries, Empi/Donjoy, Takeda Pharmaceutical Company, RS medical, Covidien, Mallinckrodt Pharmaceuticals, Reckit Benckiser Pharmaceuticals, and Neurotech. MJ is a consultant to XenoPort, Inc. RK and GS are employees of and own stock in XenoPort, Inc.

bit.ly/1jEZn9c

American Academy of Neurology 66th Annual Meeting – Pennsylvania Convention Center, Philadelphia, PA – April 26–May 3, 2014