The Efficacy of Gabapentin Enacarbil (GEn) in Adult Patients With Severe Primary Restless Legs Syndrome (RLS): Pooled Analyses From 3 Randomized Trials

I3-1.009
P5.290

Daniel O. Lee,[1] William Ondo,[2] Alon Y. Avidan,[3] Diego García-Borreguero,[4] Mark Jaros,[5] Richard Kim,[6] Gwendoline Shang[6]

[1]Baptist Health Neurology, Richmond, KY; [2]The University of Texas Medical School at Houston, Houston, TX; [3]Ronald Reagan UCLA Medical Center, Los Angeles, CA; [4]Sleep Research Institute, Madrid, Spain; [5]Summit Analytical, LLC, Denver, CO; [6]XenoPort, Inc., Santa Clara, CA

Introduction

- Gabapentin enacarbil (GEn) is an actively transported prodrug of gabapentin that has been approved by the United States FDA at a dose of 600 mg once daily for the treatment of moderate-to-severe primary RLS in adults.[1]
- In 3 randomized, double-blind, placebo-controlled studies (XP052/XP053/XP081) in patients with moderate-to-severe primary RLS, GEn 1200 mg improved RLS symptoms compared with placebo with regard to the mean change from baseline in International Restless Legs Scale (IRLS) total score and the proportion of responders on the investigator-rated Clinical Global Impression–Improvement (CGI-I) scale at week 12.[2–4]
 - In studies XP053 and XP081, similar results were obtained with GEn 600 mg.[3,4]
 - GEn was well tolerated in all 3 studies; the most commonly reported adverse events (AEs) were somnolence and dizziness.[2–4]
- The primary analyses focused on the overall study populations of patients with moderate-to-severe RLS, but did not investigate efficacy in the subset of patients with severe primary RLS.

Objective

- To further investigate the efficacy and safety of GEn in the subset of patients with severe primary RLS using pooled data from the XP052, XP053, and XP081 studies from the GEn 600 mg, GEn 1200 mg, and placebo treatment groups.

Methods

Study design

- The study designs of the three 12-week, randomized, double-blind, placebo-controlled trials XP052, XP053, and XP081 have previously been published.[2–4]
 - In XP052, GEn 1200 mg once daily was compared with placebo
 - In XP053, GEn 1200 mg and 600 mg once daily were compared with placebo
 - In XP081, the pharmacokinetics, efficacy and safety of GEn 600 mg, 1200 mg, 1800 mg, 2400 mg, and placebo were examined
- In all 3 studies, patients had a diagnosis of RLS using the IRLS Study Group criteria,[5] RLS symptoms ≥15 days during the month prior to screening (or, if on treatment, similar symptom frequency before treatment initiation), documented RLS symptoms for ≥4 of the 7 consecutive evenings/days during the baseline period, IRLS total score ≥15, and had discontinued any other RLS treatment ≥2 weeks prior to baseline.

Pooled analysis

- Patient-level data were pooled for patients receiving GEn (600 mg or 1200 mg) or placebo once daily from the XP052, XP053, and XP081 trials.
- The safety population comprised all patients who had received at least one dose or portion of a dose of study medication. Efficacy analyses were performed on the modified intent-to-treat population (MITT; all patients in the safety population who had a baseline and ≥1 postbaseline IRLS assessment).
- Analyses were based on the subset of patients with severe primary RLS (ie, those having a baseline IRLS total score ≥24) using the MITT population.
- The efficacy outcomes assessed were mean change from baseline to week 12 in IRLS total score and the proportion of responders (rated “much” or “very much” improved) on the investigator-rated CGI-I scale at week 12.
- For the change from baseline IRLS total scores, treatment effects were analyzed using a mixed-effect model for repeated measures (MMRM) with an unstructured covariance matrix, including fixed effects for visit day (categorical), treatment group, and their interaction, as well as baseline IRLS total score.
- For the percentage of CGI-I responders, treatment effects were analyzed using a logistic regression model, with treatment included as the main effect.
- Safety and tolerability, including evaluations of all AEs, were analyzed according to multiple clinical and laboratory measurements using the safety population (patients who had received ≥1 dose or portion of a dose of study treatment).

Results

Patients

- Demographic and baseline characteristics are shown in Table 1.
- In this pooled analysis, 45.1% (110/244) of patients in the placebo group, 49.7% (80/161) in the GEn 600 mg group, and 44.7% (119/266) in the GEn 1200 mg group had severe RLS (IRLS total score ≥24) at baseline (Table 1).
- Of the patients with severe RLS, 77.3% (85/110) in the placebo group, 80.0% (64/80) in the GEn 600 mg group, and 84.0% (100/119) in the GEn 1200 mg group completed their respective study.

Table 1. Baseline characteristics of the safety population of patients with severe RLS at baseline (MITT population)

Characteristic	Placebo (n=110)	GEn 600 mg (n=80)	GEn 1200 mg (n=119)
Age, years			
Mean (SD)	49 (11.7)	47 (12.2)	52 (10.4)
Sex, n (%)			
Female	72 (65.5)	50 (62.5)	78 (65.6)
Male	38 (34.6)	30 (37.5)	41 (34.5)
Race, n (%)			
White/Caucasian	104 (94.6)	74 (92.5)	115 (96.6)
Mean IRLS total score, n (%)			
Total patients with IRLS scores, n	244	161	266
<24 at baseline	134 (54.9)	81 (50.3)	147 (55.3)
≥24 at baseline	110 (45.1)	80 (49.7)	119 (44.7)
IRLS total score			
Mean (SD)	27.5 (3.0)	27.6 (3.0)	28.0 (3.3)
Duration of RLS symptoms, years			
Mean (SD)	14 (13.5)	14 (13.5)	14 (13.1)
Prior RLS treatment, n (%)			
No	57 (51.8)	52[b] (66.7)	73 (61.3)
Yes[a]	53 (48.2)	26[b] (33.3)	46 (38.7)

[a]Includes patients whose treatment terminated prior to the month before the start of study drug, who received treatment within the month of the start of study drug or within the previous month; [b]Out of 78 patients; 2 patients had missing data.
GEn: gabapentin enacarbil; IRLS: International Restless Legs Scale; MITT: modified intent-to-treat; RLS: restless legs syndrome; SD: standard deviation

Efficacy

- The LS mean (SE) change from baseline IRLS total score at week 12 was greater for GEn 600 mg and 1200 mg compared with placebo; these differences were statistically significant (Figure 1).

Figure 1. LS mean changes (SE) and treatment differences in IRLS total score from baseline at week 12

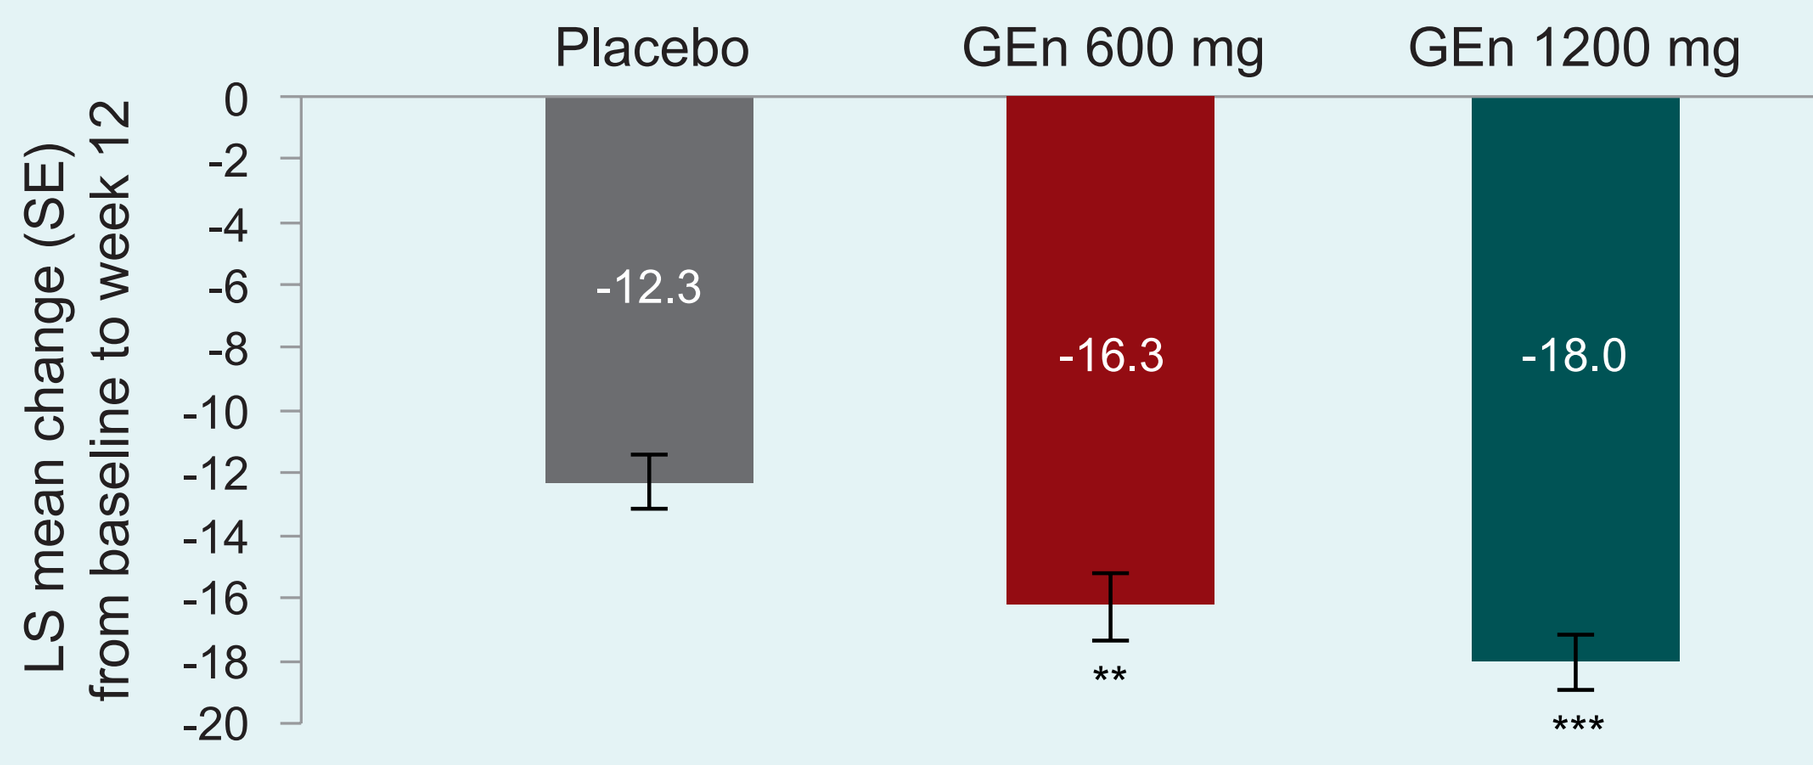
Placebo
GEn 600 mg
GEn 1200 mg
LS mean change (SE) from baseline to week 12
-12.3
-16.3
-18.0
**

Mean treatment difference (SE):
GEn 600 mg vs placebo: -4.0 (1.4); P=0.003
GEn 1200 mg vs placebo: -5.7 (1.2); P<0.001

IRLS: International Restless Legs Scale; GEn: gabapentin enacarbil; LS: least squares; SE: standard error

- Compared with placebo, GEn 600 mg and GEn 1200 mg significantly improved IRLS total score from baseline at each time point, starting as early as week 1 (Figure 2).
- Although this analysis was not powered to detect differences between GEn doses, statistically significant changes from baseline in IRLS total score were observed with GEn 1200 mg vs 600 mg at weeks 3, 4 and 6; for other time points, the changes from baseline were numerically greater with GEn 1200 mg without statistical significance.

Figure 2. LS mean changes (SE) in IRLS total score from baseline by visit

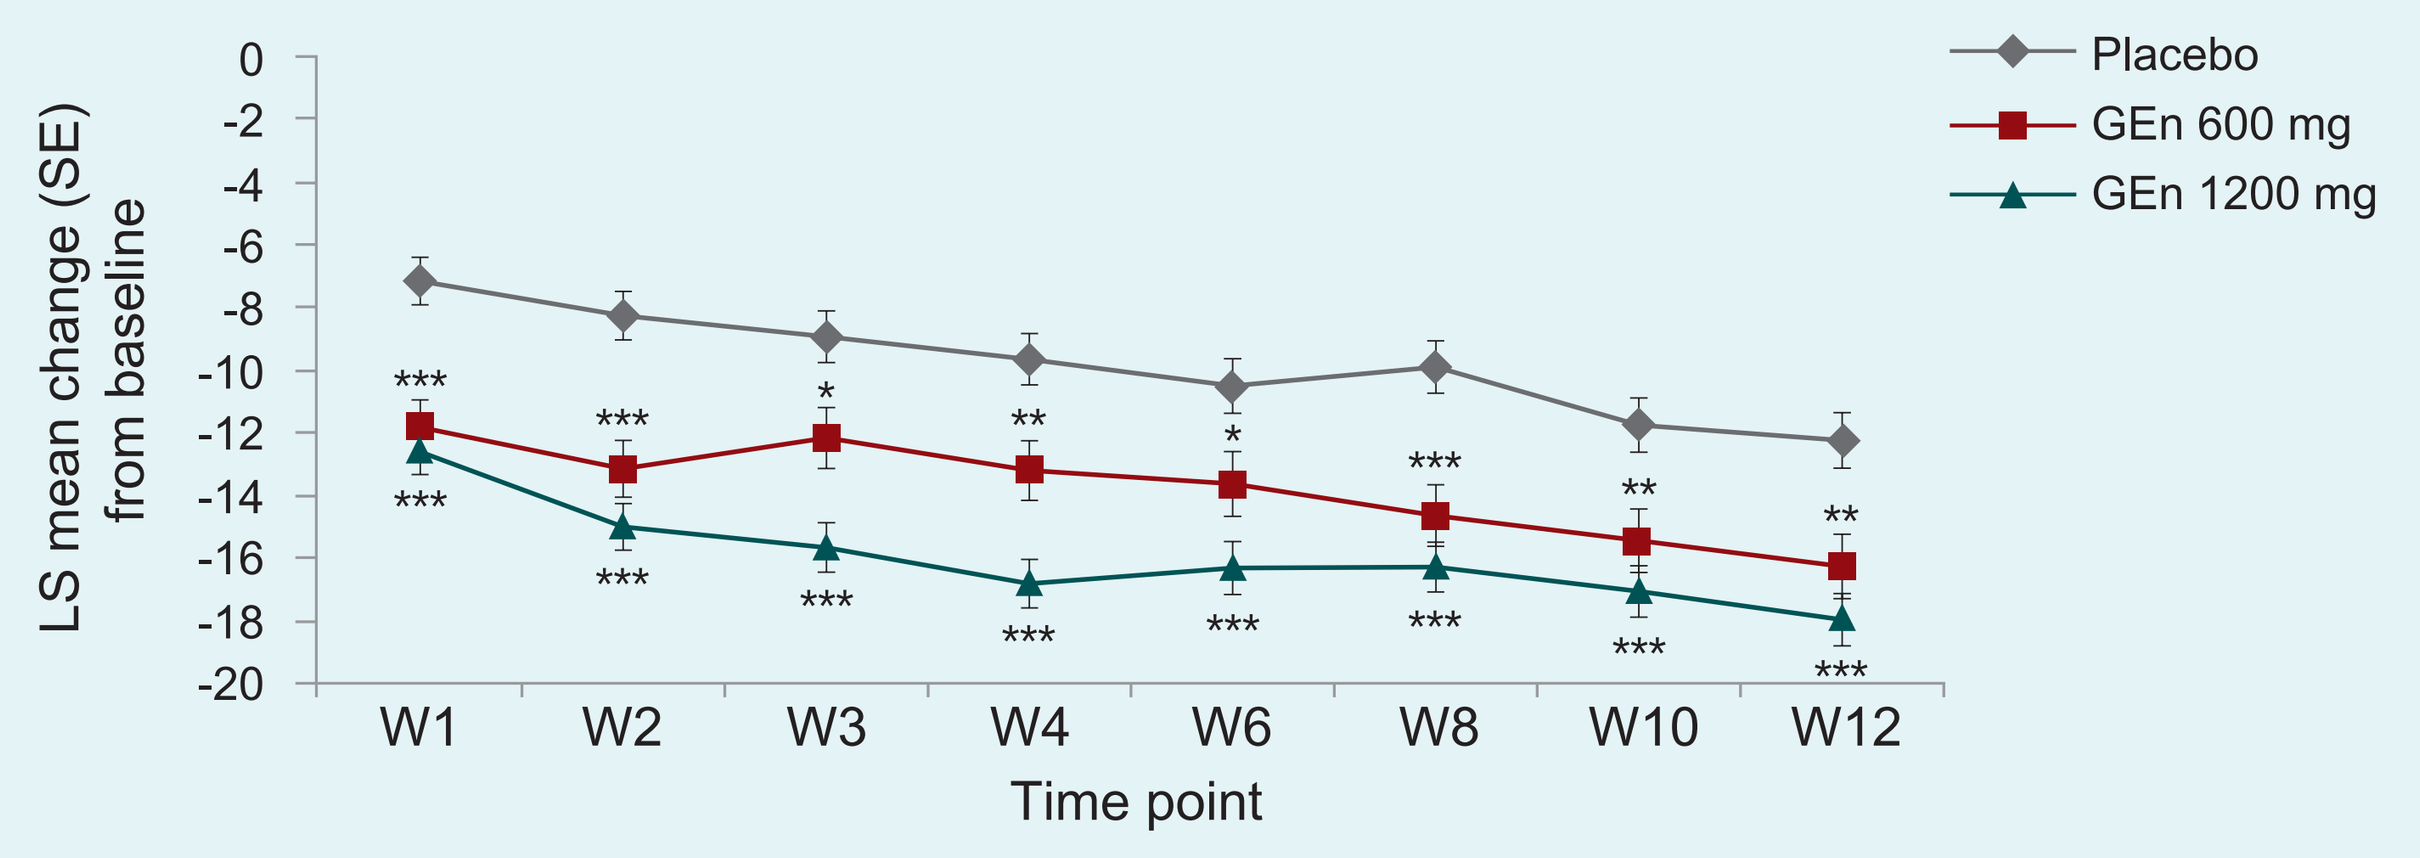
Placebo
GEn 600 mg
GEn 1200 mg
LS mean change (SE) from baseline
W1
W2
W3
W4
W6
W8
W10
W12
Time point

*P<.05; **P<.01; ***P<.001
P values are for pairwise comparisons of the differences in treatments at each time point (GEn 600 mg vs placebo and GEn 1200 mg vs placebo)
GEn: gabapentin enacarbil; LS: least squares; W: week

- Compared with placebo, significantly more patients were responders according to the investigator-rated CGI-I at each time point with GEn 600 mg and 1200 mg (Figure 3). At week 12, patients treated with GEn 600 mg or 1200 mg were significantly more likely than placebo-treated patients to be responders on the CGI-I (Table 2).

Figure 3. Percentage of responders (“much” or “very much” improved) on the investigator-rated CGI-I by visit

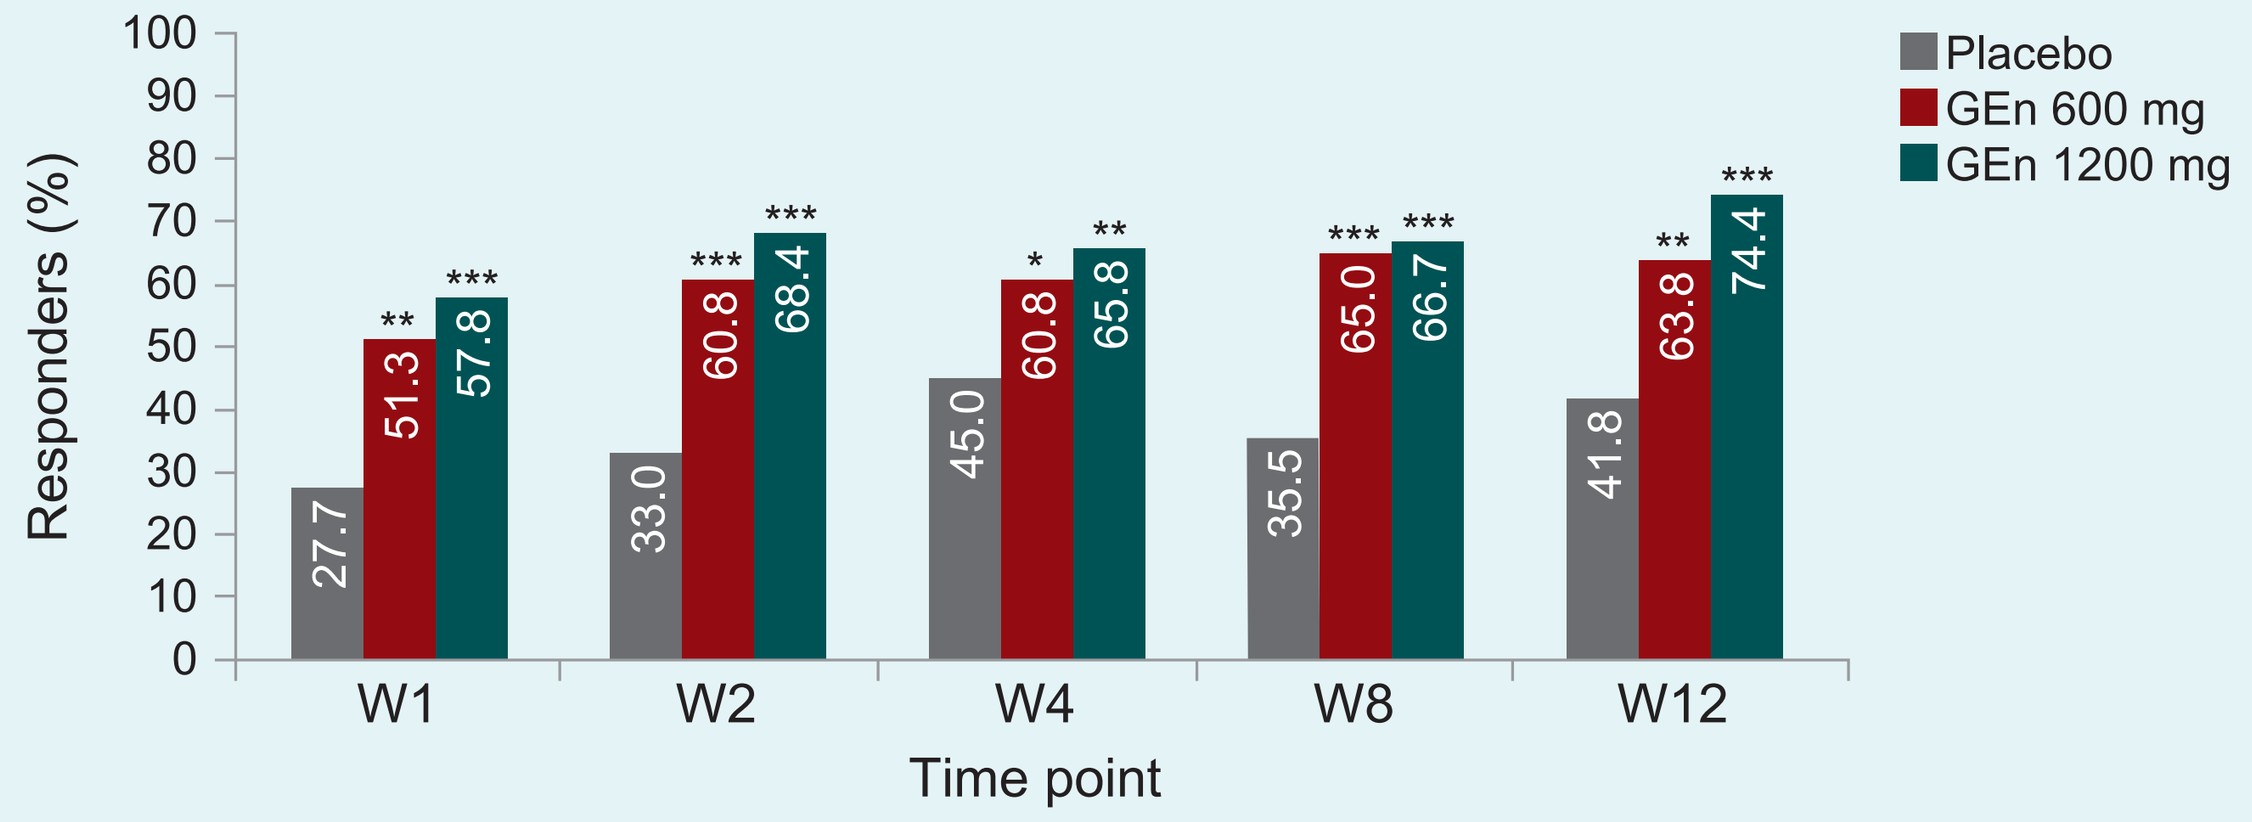
Placebo
GEn 600 mg
GEn 1200 mg
Responders (%)
27.7
51.3
57.8
33.0
60.8
68.4
45.0
60.8
65.8
35.5
65.0
66.7
41.8
63.8
74.4
W1
W2
W4
W8
W12
Time point

*P<.05; **P<.01; ***P<.001
P values are for GEn 600 mg vs placebo or GEn 1200 mg vs placebo
CGI-I: Clinical Global Impression-Improvement; GEn: gabapentin enacarbil; W: week

Table 2. Percentage of responders (“much” or “very much” improved) and odds ratios on the investigator-rated CGI-I at week 12

	Placebo (n=110)	GEn 600 mg (n=80)	GEn 1200 mg (n=117)
Responders, %	41.8	63.8	74.4
OR (95% CI) vs placebo	–	2.4 (1.4–4.4)	4.0 (2.3–7.1)
P value	–	.003	<.001

CGI-I: Clinical Global Impression-Improvement; CI: confidence interval; GEn: gabapentin enacarbil; OR: odds ratio

Tolerability

- The most commonly reported treatment-emergent AEs with GEn 600 mg and 1200 mg in patients with severe primary RLS were somnolence and dizziness (Table 3).

Table 3. Most frequent treatment-emergent adverse events reported in ≥5% of the safety population

Adverse event, n (%)	Placebo (n=111)	GEn 600 mg (n=81)	GEn 1200 mg (n=119)
Any adverse event	81 (73.0)	68 (84.0)	99 (83.2)
Somnolence	3 (2.7)	17 (21.0)	28 (23.5)
Headache	13 (11.7)	10 (12.3)	14 (11.8)
Dizziness	3 (2.7)	11 (13.6)	22 (18.5)
Nausea	7 (6.3)	7 (8.6)	9 (7.6)
Fatigue	6 (5.4)	7 (8.6)	9 (7.6)
Nasopharyngitis	5 (4.5)	6 (7.4)	9 (7.6)
Diarrhea	7 (6.3)	3 (3.7)	3 (2.5)
Constipation	6 (5.4)	2 (2.5)	6 (5.0)
Insomnia	2 (1.8)	5 (6.2)	4 (3.4)
Irritability	1 (0.9)	3 (3.7)	6 (5.0)
Paresthesia	6 (5.4)	1 (1.2)	0 (0)
Sinusitis	0 (0)	5 (6.2)	2 (1.7)

GEn: gabapentin enacarbil

Conclusions

- This is the first analysis of GEn efficacy in the subset of patients with severe primary RLS, and provides evidence that even patients with severe cases of RLS can benefit from treatment with GEn.
- In this pooled analysis of patients with severe primary RLS from 3 randomized trials, treatment with GEn was associated with statistically significant improvements in response compared with placebo.
 - At week 12, change from baseline IRLS total score was statistically significantly greater with GEn 600 mg and 1200 mg compared with placebo. Statistically significant treatment differences emerged as early as week 1 and were maintained throughout the treatment phase.
 - At week 12, there was a significantly greater proportion of responders according to the investigator-rated CGI-I.
- Similar to the AEs seen for the overall study populations,[2–4] the most common TEAEs in patients with severe RLS were somnolence and dizziness. The AEs reported in this study are consistent with the overall safety profile of GEn.[1]
- According to product labeling, the recommended dose in adult patients with moderate-to-severe primary RLS is 600 mg once daily. A daily dose greater than 1200 mg provided no additional benefit, but caused an increase in adverse reactions.
- In considering best treatments for adult patients with severe primary RLS, careful consideration should be given to the benefits and risks of dose selection for these patients.
- Consistent with published data in the overall moderate-to-severe RLS patient population, GEn 600 mg and 1200 mg demonstrated consistent and significant efficacy compared with placebo in these patients with severe RLS.

References

1. Horizant [Prescribing Information]. Santa Clara, CA: XenoPort, Inc; 2013.
2. Kushida C, et al. *Neurology*. 2009;72:439–446.
3. Lee D, et al. *J Clin Sleep Med*. 2011;7:282–292.
4. Lal R, et al. *Clin Neuropharmacol*. 2012;35:165–173.
5. Allen R, et al. *Sleep Med*. 2003;4:101–119.

Acknowledgments

These studies and this analysis were conducted by XenoPort, Inc., Santa Clara, CA.
Financial support for medical writing was provided by XenoPort, Inc.

Disclosures

DL is a speaker and consultant for XenoPort, Inc. WO was a speaker for UCB Pharma, Lundbeck, Merz, Ipsen, and TEVA, and received grant support from US World Meds, UCB Pharma, and Ipsen. AYA served as a consultant for UCLA. DGB served as a consultant for UCB, XenoPort, Inc., Impax Pharmaceuticals, and Otsuka. MJ is a consultant to XenoPort, Inc. RK and GS are employees of and own stock in XenoPort, Inc.

bit.ly/1jEZn9c

American Academy of Neurology 66th Annual Meeting – Pennsylvania Convention Center, Philadelphia, PA – April 26–May 3, 2014